Population Pharmacokinetic/Pharmacodynamic Modeling of a next generation recombinant human Factor VIIa (LR769) to Derive the Dose to be Studied in Phase 3

Authors: Jules Heuberger[1]*, Johan Frieling[2], Matthijs Moerland[1], Joannes Reijers[1], Koos Burggraaf[1], Cornelis Kluft[3], Jean-Francois Schved[4], Jasper Stevens[1]

[1]Centre for Human Drug Research, Leiden, the Netherlands; [2]LFB USA, Framingham, MA, USA; [3]GBS, Leiden, the Netherlands; [4]CHU Montpellier, Montpellier, France. *Presenting author

INTRODUCTION

Recombinant human Coagulation Factor VIIa (rhFVIIa, LR769) is being developed for the treatment of bleeding episodes in hemophilia A or B patients.

AIM

To develop a population PK/PD model to characterize the Factor VIIa concentration-effect relationship for activated partial thromboplastin time (aPTT), thromboelastography (MCF: maximum clot firmness) and Prothrombin fragments 1+2 (F1+2). This model was then used to optimize a treatment regimen that is expected to be effective in treating and preventing bleedings in hemophilia A/B patients with inhibitors.

METHODS

rhFVIIa concentration data (343 data points) and its effects on PD blood markers (359 data points for aPTT and 240 data points for F1+2 and MCF) after the administration of 25, 75 and 225 µg/kg LR769 to 15 hemophilia A/B patients from a randomized, open label multiple dose cross-over study were used to develop a PK/PD model using the FOCE with interaction estimation method in NONMEM® 7.2. FVIIa activity was assessed by modified STACLOT rTF assay. The identified population PK/PD models were used to simulate the response curves as a function of Factor VIIa activity with different dosing regimens.

RESULTS

Using a stepwise approach, a two-compartment model for bolus IV administration resulted in the best model fit to describe the PK of rhFVIIa, with lean body mass (LBM) as a covariate on Vd and inter-individual variability on the elimination rate constant and a proportional error model. Using the PK model as a driving factor, the same approach identified the three optimal models describing the different PD measures. A sigmoidal maximal effect model with proportional error was identified for MCF, F1+2 (increasing with increasing FVIIa) and aPTT (decreasing with increasing FVIIa) with the latter two having a gamma fixed at 1. The effect in F1+2 showed a delay, which was modelled using an effect compartment (rate constant KE0). IOV or IIV was identified on the baseline level (E0) for all three parameters, IIV on the Emax for aPTT and on the EC50 and KE0 for F1+2. Several dose regimens were simulated and evaluated for desired effect levels from these models.

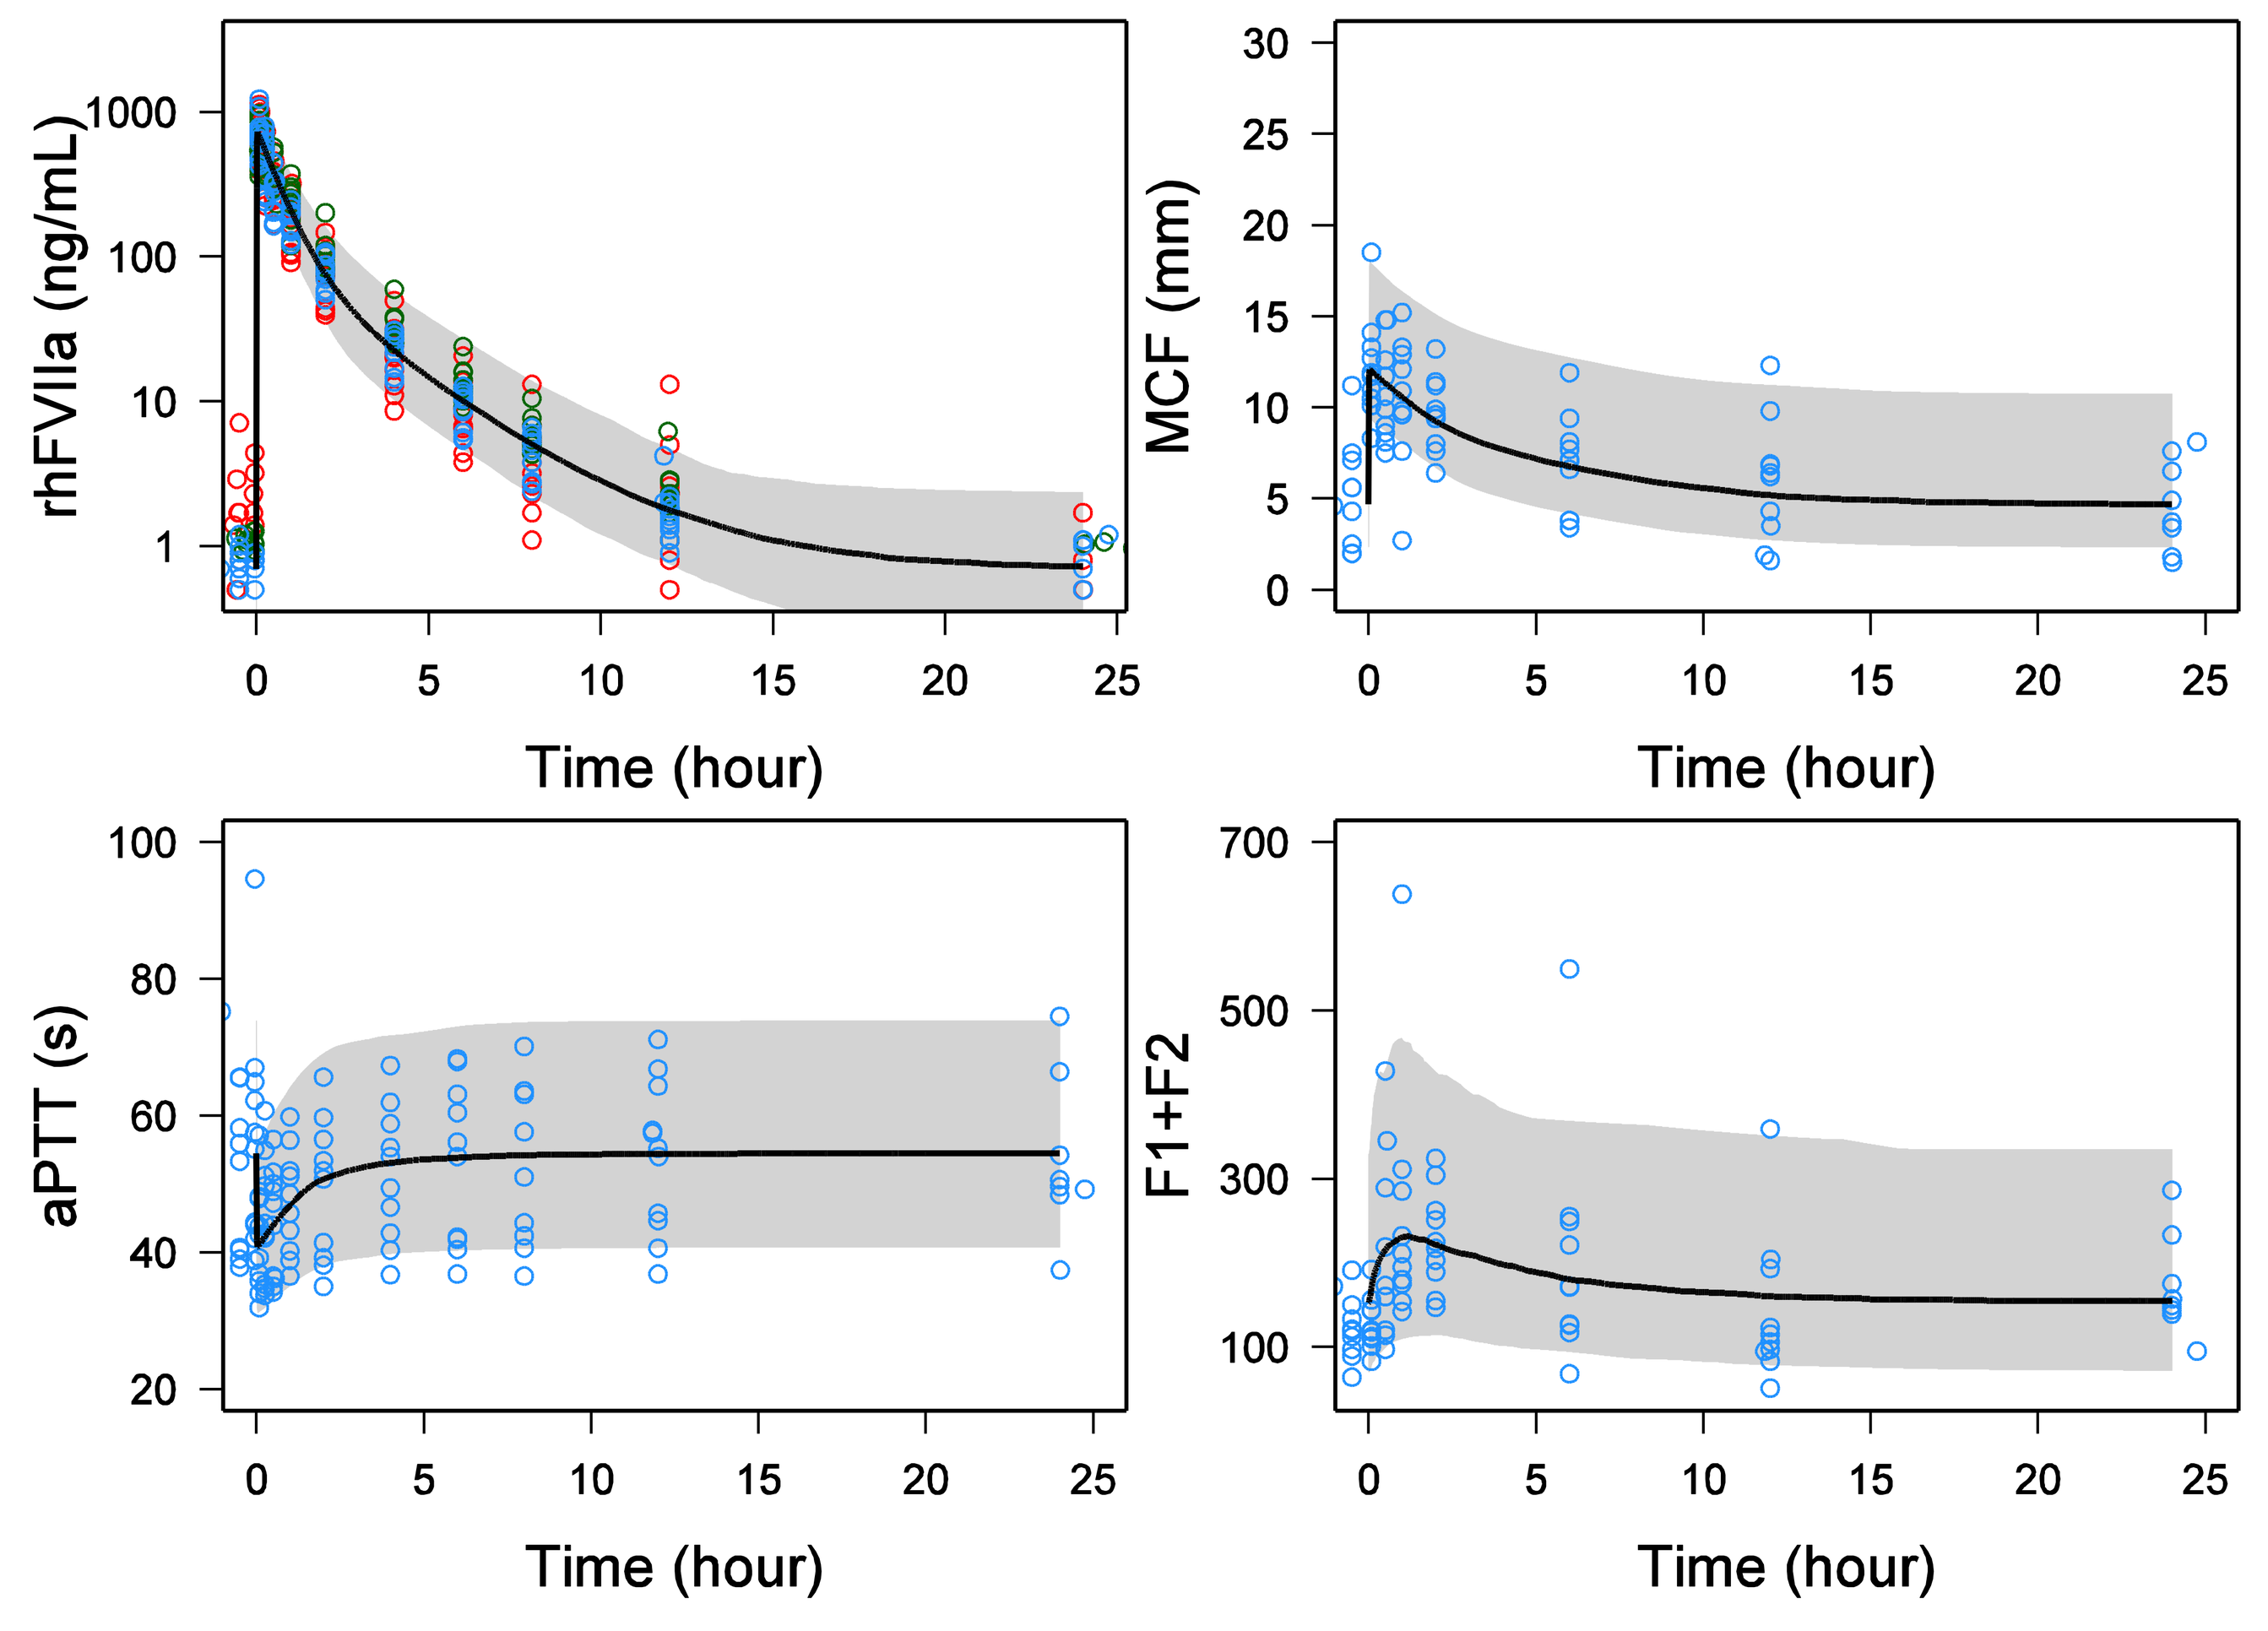
rhFVIIa (ng/mL)
MCF (mm)
aPTT (s)
F1+F2
Time (hour)

Figure 2: Visual Predictive Check of 75 µg/kg LR769. Blue (75), red (25) and green (225) circles: observations corrected for 75 µg/kg dose; Black line, typical predicted concentration; Gray area, 95% prediction interval.

RESULTS continued

	aPTT in s		MFC in mm		F1+2 in pmol/L		
		IIV		IOV		IOV	IIV
E0 (s, mm, pmol/L, resp.)	55.1 (4)	14	3.32 (12.8)	58	148 (7.7)	39	-
Emax (s, mm, pmol/L, resp.)	21.5 (13.0)	14	11 (17)	-	863 (40.8)	-	-
EC50 (ng/mL)	380 (25.9)	-	87.4 (80.9)	-	1280 (52.9)	-	46
Gamma (-)	1 fix	-	0.47 (17.6)	-	1 fix	-	-
KE0 (/h)	-	-	-	-	0.333 (28.2)	-	101
Proportional error	0.0116	-	0.0888	-	0.0625	-	-

Table 1: Median parameter estimates (RSE in %) for all PD measures

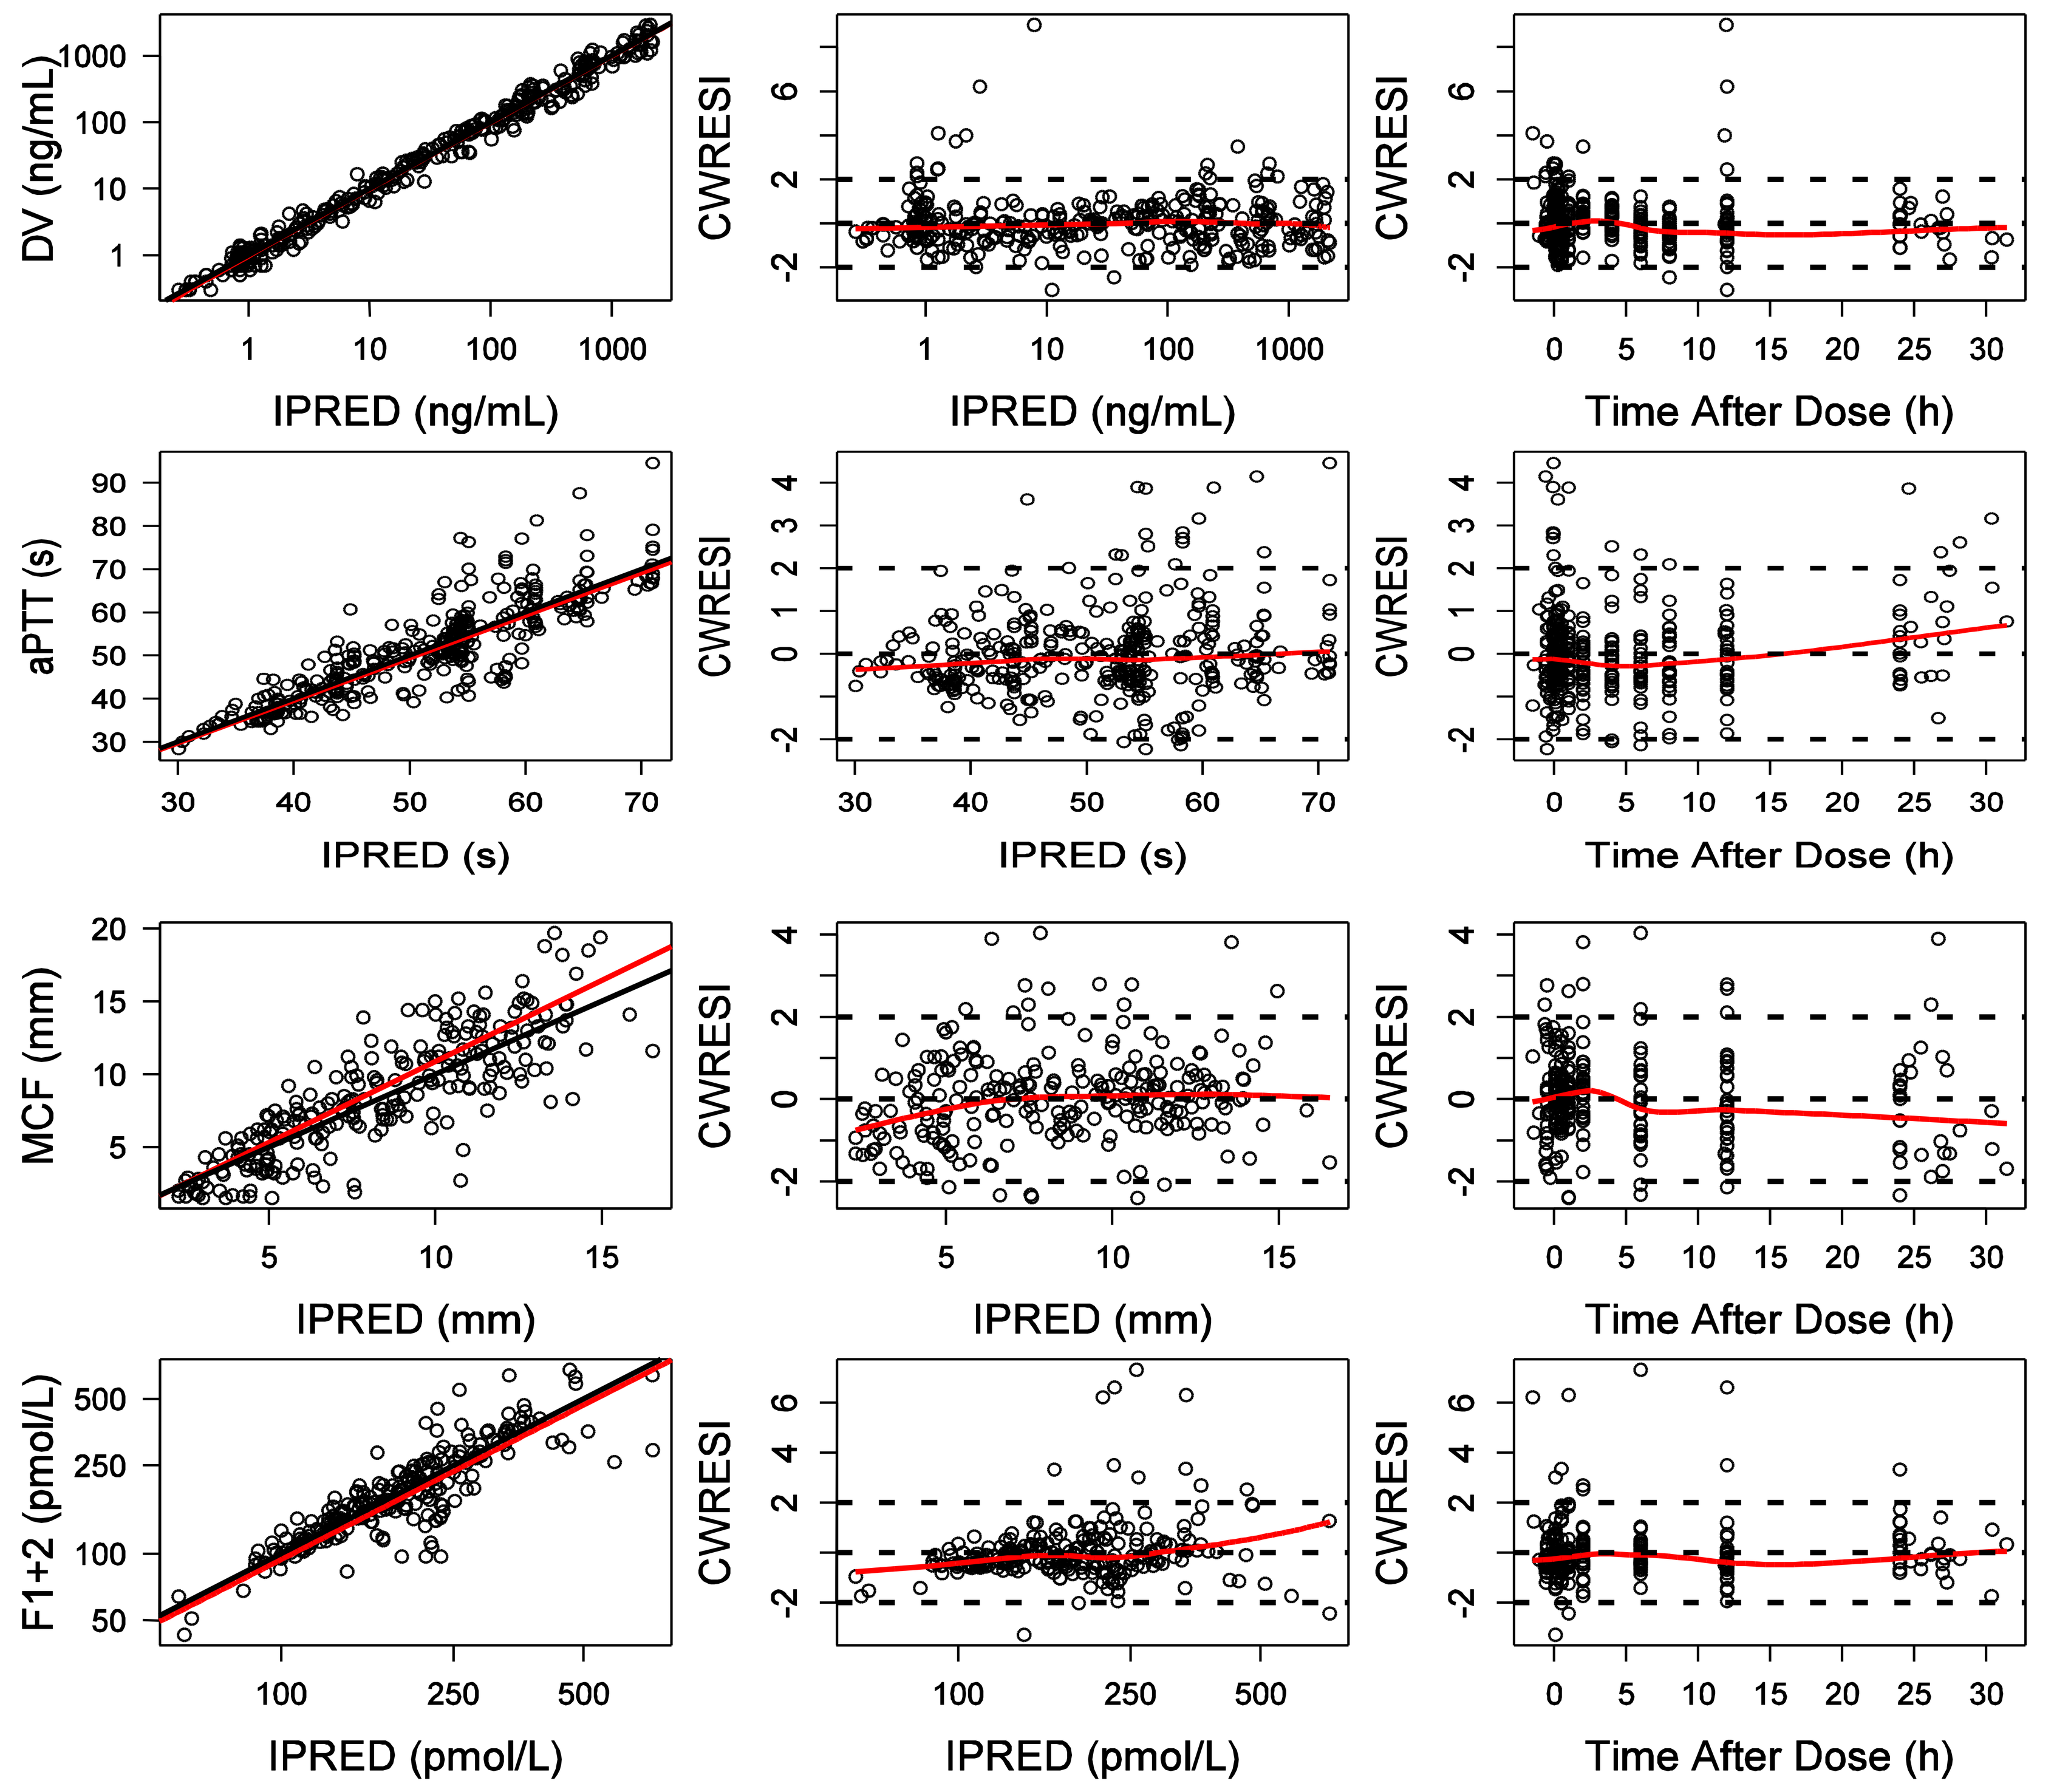
DV (ng/mL)
CWRESI
IPRED (ng/mL)
Time After Dose (h)
aPTT (s)
IPRED (s)
MCF (mm)
IPRED (mm)
F1+2 (pmol/L)
IPRED (pmol/L)

Figure 1: Diagnostic plots PK and PK/PD models. Red line; regression (left) or LOESS line (middle and right, span=0.75) through the observations.

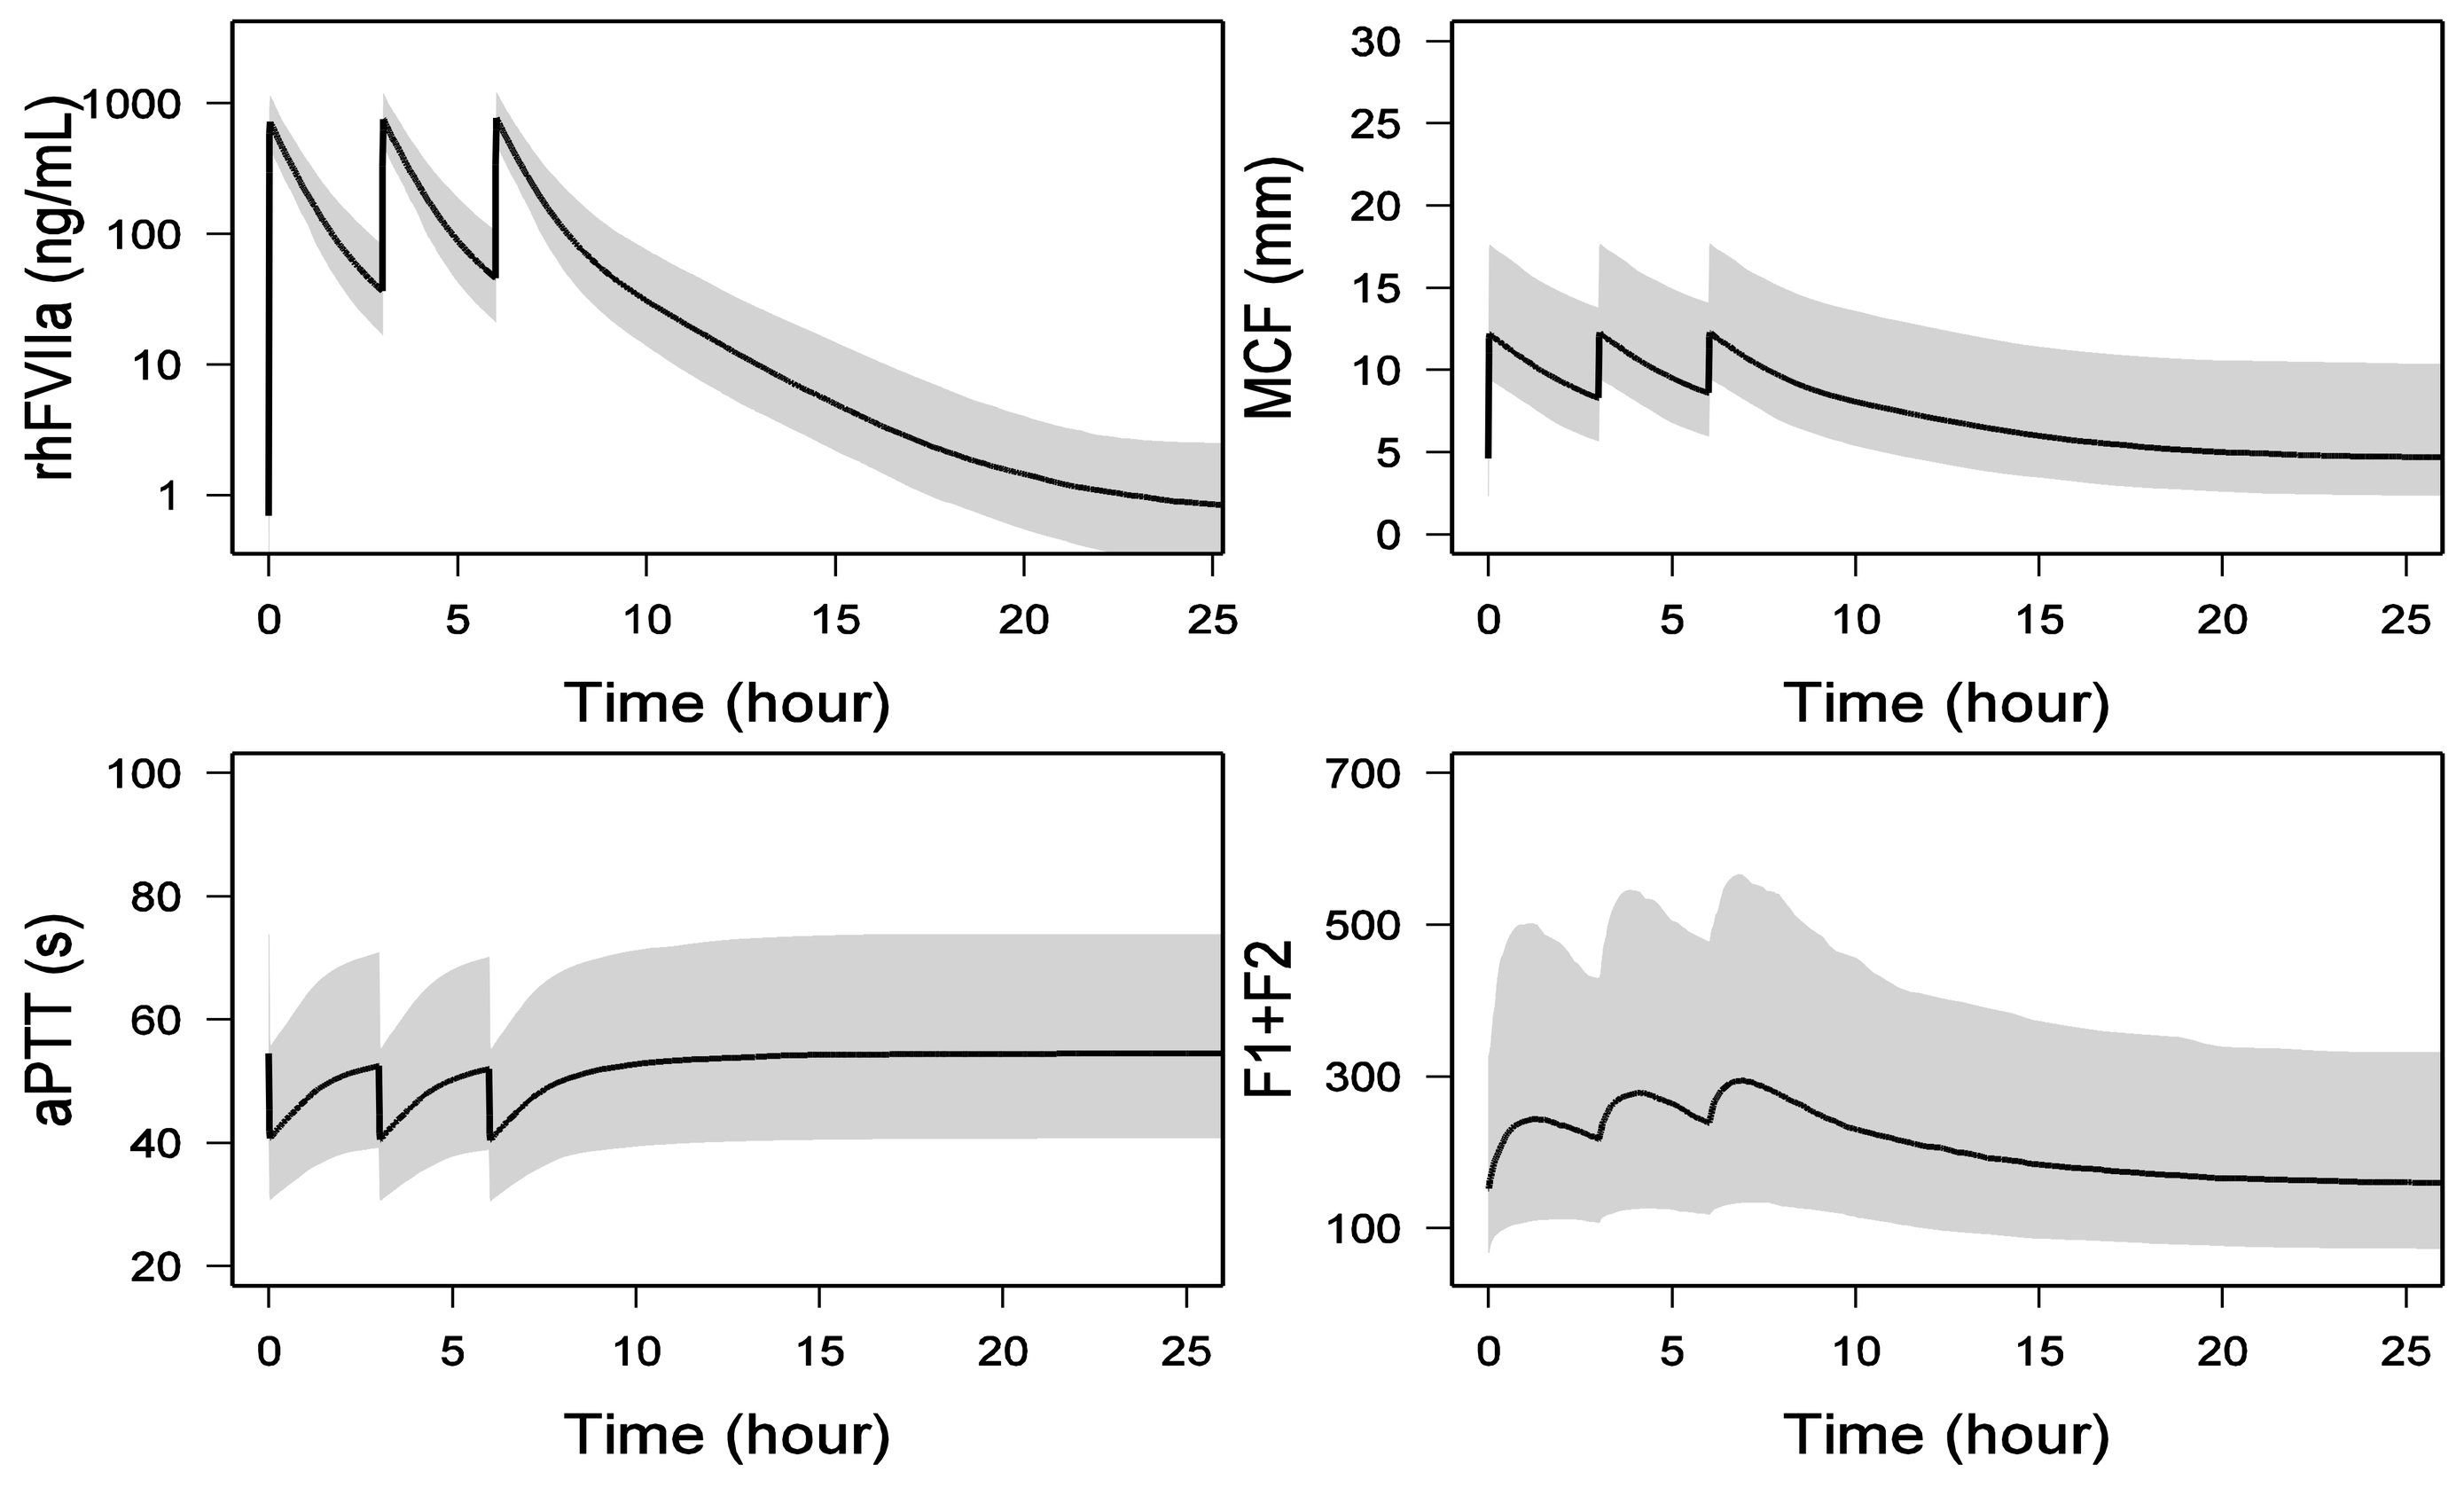
rhFVIIa (ng/mL)
MCF (mm)
aPTT (s)
F1+F2
Time (hour)

Figure 3: Simulations of 75 µg/kg rhFVIIa dose 3 times every 3 hours and its effects. Black line, typical predicted concentration; Gray area, 95% prediction interval.

CONCLUSIONS

- **LR769 shows linear PK over the dose-range studied**
- **The concentration-effect relationship of LR769 can be accurately described**
- **Simulated optimal dose regimens seems to be 75 µg/kg every 3 hours, or 225 µg/kg if needed followed 9 hours later by 75 µg/kg**

Preliminary results of the ongoing Phase 3 study indicate the predicted dose regimens may be effective and safe

Centre for Human Drug Research | Zernikedreef 8 | 2333 CL Leiden | The Netherlands
Tel +31 71 52 46 400 | info@chdr.nl | www.chdr.nl